Formation and Impact of the RIDE (Research Inclusion, Diversity and Equity) Council to Promote Diverse Engagement in MS Research

H. Schmidt[1], J. Caines[1], D. V. Cavalier[1], S. Feliciano[1], T. Johnson[1], A. A. Williams[1], S. Ngorsuraches[2];
[1]Accelerated Cure Project, Waltham, MA, [2]Auburn University, Auburn, AL.

Background and Aims

There is an increasing awareness of the lack of diversity in MS research participation and knowledge about how this disparity perpetuates inequities in MS healthcare. However, awareness and knowledge are not enough to resolve long-standing imbalances in research representation.

Accelerated Cure Project (ACP) is a research-focused patient advocacy organization and the sponsor of the iConquerMS people-powered research network which has enrolled over 8,500 individuals with MS since being established in 2014. The network includes people from many backgrounds, identities, and cultures but, similar to most MS research cohorts, has never fully reflected the diversity of the MS population.

To address this imbalance, iConquerMS leaders created a council of people affected by MS called the iConquerMS RIDE (Research Inclusion, Diversity and Equity) Council. Its goal is to promote diversity and inclusion in iConquerMS and other MS research efforts through collaboration with people affected by MS from underrepresented communities.

Methods

In the fall of 2021, ACP hired a program manager and established a steering committee purposely including people affected by MS and researchers from diverse backgrounds. In January 2022, the committee sent an email invitation to iConquerMS members explaining the goals of the RIDE Council and its plans for operation including monthly virtual meetings and co-creation of strategies and plans.

Interested individuals were directed to an application where they could describe their connection to MS, their experience with MS advocacy and/or research, and the communities to which they belong that are underrepresented in MS research. Applications were reviewed by the steering committee.

Results

In all, 54 applications were received from iConquerMS members belonging to underrepresented communities. The steering committee selected 28 applicants and formed three sub-councils based on their identities:

Black/African-American: 12 members

Hispanic/LatinX: 8 members

Emerging voices (Native American, Asian-American, LGBTQ+, rural, veteran): 8 members

Two orientation sessions were held in March 2022 to provide background and context, create a set of community agreements, and discuss council operations. Following that, the Council began meeting on a monthly basis, primarily in sub-council meetings but occasionally as a full council.

Each sub-council has led efforts to increase diverse participation in iConquerMS and support other needs of their communities. For example, each sub-council designed a distinct tri-fold brochure describing iConquerMS for their communities; the Black/African-American sub-council created a series of iConquerMS-branded infographics providing tailored advice about managing MS; and the Hispanic/LatinX sub-council is guiding efforts to create a Spanish-language version of iConquerMS.

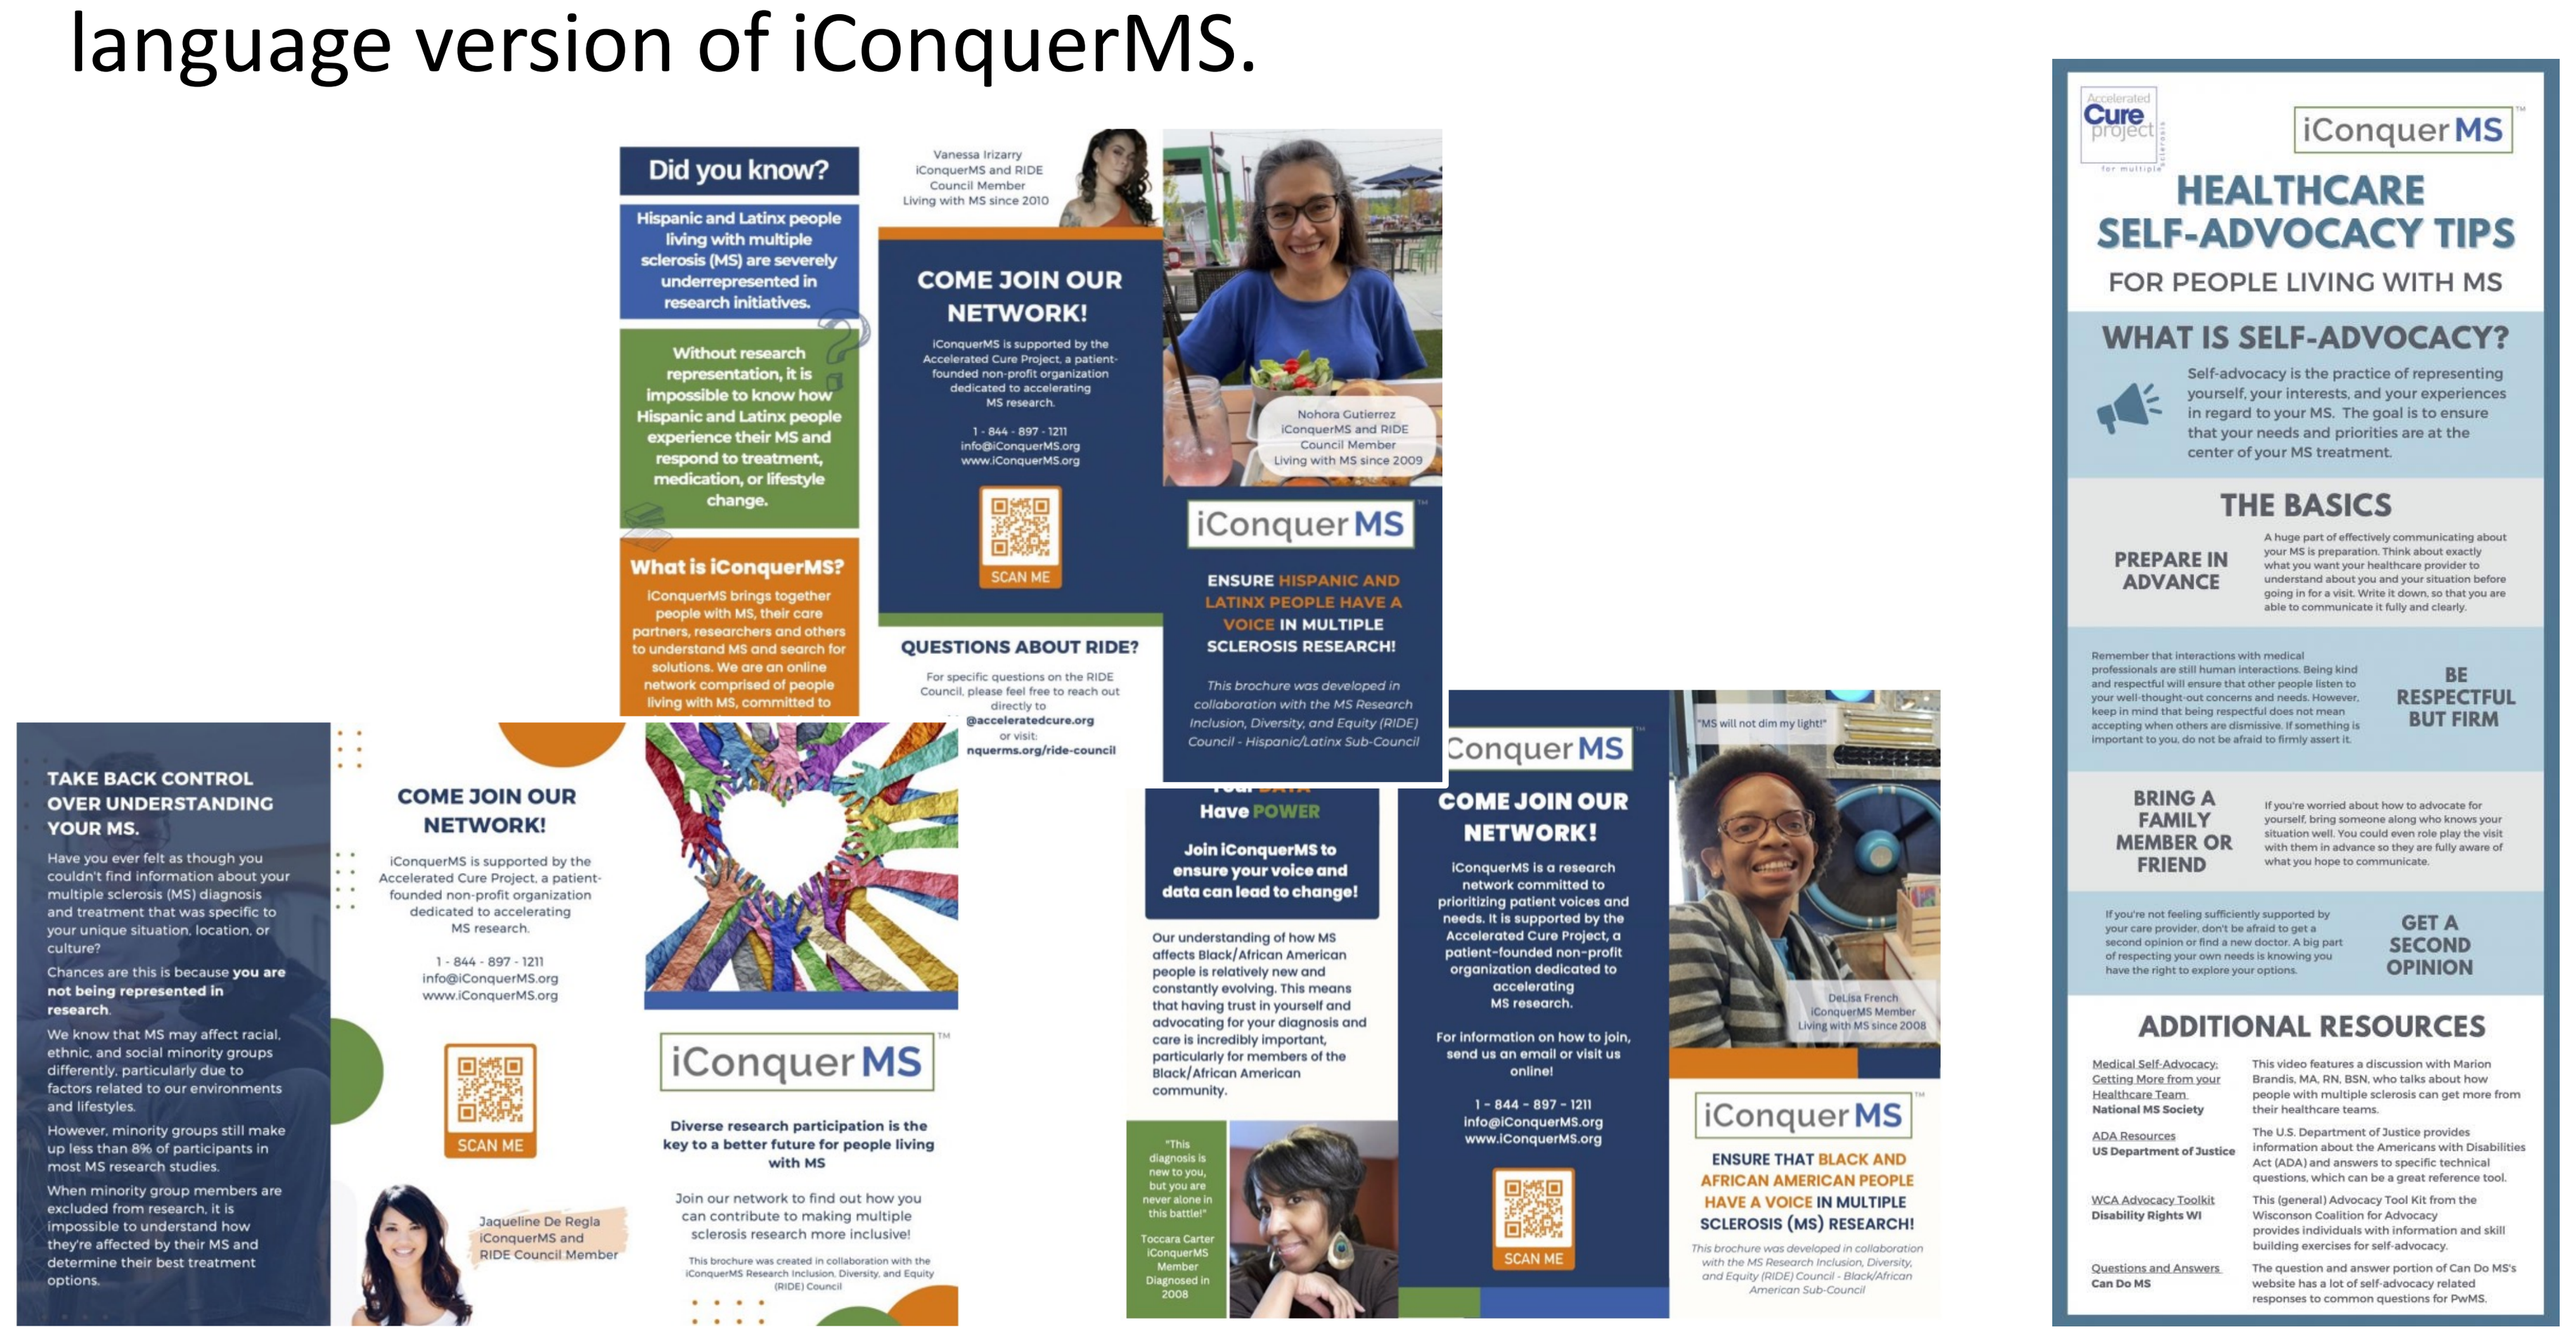
Did you know?
COME JOIN OUR NETWORK!
What is iConquerMS?
QUESTIONS ABOUT RIDE?
TAKE BACK CONTROL OVER UNDERSTANDING YOUR MS.
HEALTHCARE SELF-ADVOCACY TIPS
FOR PEOPLE LIVING WITH MS
WHAT IS SELF-ADVOCACY?
THE BASICS
PREPARE IN ADVANCE
BE RESPECTFUL BUT FIRM
BRING A FAMILY MEMBER OR FRIEND
GET A SECOND OPINION
ADDITIONAL RESOURCES

Figure 1: Examples of materials co-created with RIDE sub-councils

Recognizing that other MS research efforts could benefit from the experience and insights offered by RIDE Council members, we have facilitated discussions and engagements between RIDE Council members and MS researchers. Examples include:

- Discussions and review of materials to improve the cultural relevance of a survey on barriers to healthcare access
- Focus group sessions to provide insights on the wording of specific patient-reported outcomes intended for use in MS clinical trials
- Evaluation of psychotherapy materials translated from another language for cultural fit prior to their use in a clinical trial
- Guidance of the development of workshops to obtain input on a digital health application from racial/ethnic minority populations

Conclusions

We have demonstrated that people affected by MS belonging to underserved populations are enthusiastic about increasing representation of their groups in MS research, and are eager to share their expertise with researchers. Formation of a standing council to facilitate collaboration among these communities and researchers is an effective approach that promotes trust-building and a sense of shared mission. Further work and support are needed to sustain and expand the council's efforts to promote greater diversity and inclusion in MS research.

Acknowledgements

Funding for the RIDE Council has been provided by Bristol Myers Squibb, Biogen, Novartis, Genentech, and Sanofi.

Disclosures

The authors have no conflicts of interest to disclose.

For more information

Please contact Jordan Caines (jcaines.acp@gmail.com) for more information about the RIDE Council.

© Poster Template by Genigraphics® 1.800.790.4001 www.genigraphics.com